Development of Robust Orally Dispersible Powder Formulations

Graeme Macleod, Hanumantharaja, Brian Wilson, and Girish BV

SPI Pharma Inc, Rockwood Office Park, 503 Carr Road, Suite 210, Wilmington, DE 19809 United States • gmacleod@spipharma.com

INTRODUCTION

Despite an increased emphasis from regulators regarding the development of flexible patient oriented palatable medicines for children there remains a lack of significant commercially successful products on the market. One approach that is poorly characterized and not utilized extensively yet is that of orally dispersible powders (ODPs). This dose form offers many benefits for both formulation development as well as providing a positive patient experience with quick dosing. For the patient, an ODP enables the medicine to be taken on demand without the need for water and can be added to food if needed to aid compliance. An ODP can be quickly developed to give dosing flexibility with the same API at a wide range of dose strengths, including dosing up to 1000 mg. It can also be designed to maximize organoleptics such as taste, mouthfeel and flavorings by careful utilization of excipients, taste masking technology and other formulation additives. The objectives of this study were to evaluate the content uniformity, organoleptics and taste of a paracetamol ODP (Actimask® APAP) formulation based on the proprietary taste masked technology of SPI Pharma.

MATERIALS AND METHODS

Four separate ODP formulations were developed and further optimized based on taste assessment. These formulations had different ratios of APAP to carrier, dose of APAP (250 and 500 mg) and carrier used (Pharmasperse® and Mannogem® 2080). The blend of materials was sifted through a #20 ASTM sieve before being blended in a V-blender rotating at 12 rpm for 60 minutes. The particle size distribution of the formulations was determined using a Microtrac system. These blends were then packed in to stick packs using a development stick pack filling equipment to a target fill weight of 750 mg and 1100 mg for 1:2 and 1:1 APAP: Carrier ratio respectively and assessed for content uniformity, filling weight variation and organoleptics. Figure 1 details the various relevant parts of the equipment and process of filling the stick packs. Figure 2 shows the configuration of the hopper that relies on gravity feeding to fill the dies that in turn control the fill weight. Final ODP Formulations are given in Table 1. Each of the optimized formulations were also tested for drug release using 900 mL pH 5.8 phosphate buffer as the dissolution media, in a USP II apparatus, with paddle, at 50 RPM. The optimized formulas were also assessed for taste and organoleptics by 5 volunteers. Results for particle size and weight and content uniformity are given in Tables 2, 3, 4. The results of the organoleptic assessment are given in Table 5.

RESULTS AND DISCUSSION

Results show that the ODP carrier choice is critical both in terms of achieving the necessary technical robustness (flow, weight variation, and, content uniformity) but also in terms of the organoleptics of the final product. The results in Table 5 highlight some slight differences in the organoleptic profile of the different formulations. The biggest technical consideration for the formulator is that of trying to optimize the particle size distribution of the formulation. In this specific case where the taste masked API had a d50 of 410 microns, we showed that different formulations based on different carrier systems and drug loadings impacted significantly on the content uniformity achieved.

While both formulations were within limits, significant differences in the content uniformity achievable were seen, with the formulation that had the higher % of fines (Mannogem 2080 250 mg) having the poorest content uniformity most likely caused by segregation during the blending process. In addition to the technical performance we also found that the formulation based on Pharmasperse tended to have better overall organoleptic profile (creamy mouthfeel) able to mask the expected grittiness of the large particle sized taste masked API particles. All formulations had acceptable dissolution.

Table 1. Optimized ODP Formulations

Ingredient	Drug: Diluent 1:2 (mg/sachet)	Drug: Diluent 1:1 (mg/sachet)
Actimask Paracetamol	264*	528**
Pharmasperse or Mannogem 2080	471	544.5
Sucralose	5.6	11
Citric Acid Anhydrous	3.75	8.25
Strawberry Flavor	5.6	8.25
Total	750	1100

**equivalent to 250mg paracetamol*
*** equivalent to 500 mg paracetamol*

Table 2. Particle Size Data (μm) for the 4 ODP Formulations

Formulation	d10	d50	d90
Pharmasperse 500mg	247	343	445
Pharmasperse 250mg	242	344	449
Mannogem 2080 500mg	238	348	459
Mannogem 2080 250mg	186	326	413

Table 3. Particle Size Data (μm) for the Raw Materials

Material	d10	d50	d90
Pharmasperse	207	443	891
Mannogem 2080	75	304	598
Actimask Paracetamol	281	410	596

Table 4. Content Uniformity and Weight Variation Results for Different Formulations

Carrier	APAP Dose (mg)	Ratio APAP to Carrier	Content Uniformity (AV) of Stickpack (limit is 15)	Stickpack Fill Weight Range mg (% RSD)
Pharmasperse	250	1 to 2	5.5	742.5-765.5 (1.1)
Pharmasperse	500	1 to 1	6.4	1021.2-1092.2 (0.8)
Mannogem 2080	250	1 to 2	12.8	748.2-768.1 (0.9)
Mannogem 2080	500	1 to 1	4.8	1098.3-1129.3 (0.9)

Table 5. Summary of Feedback on Organoleptic Properties of Different ODP Formulations

Formulation	Organoleptic Assessment	Comment
Pharmasperse 2:1	++++	No after taste; creamy mouthfeel
Pharmasperse 1:1	++++	No after taste; creamy mouthfeel
Mannogem 2080 2:1	+++	No after taste; slightly gritty mouthfeel
Mannogem 2080 1:1	+++	No after taste; slightly gritty mouthfeel

Figure 1. Stickpack Filling Equipment

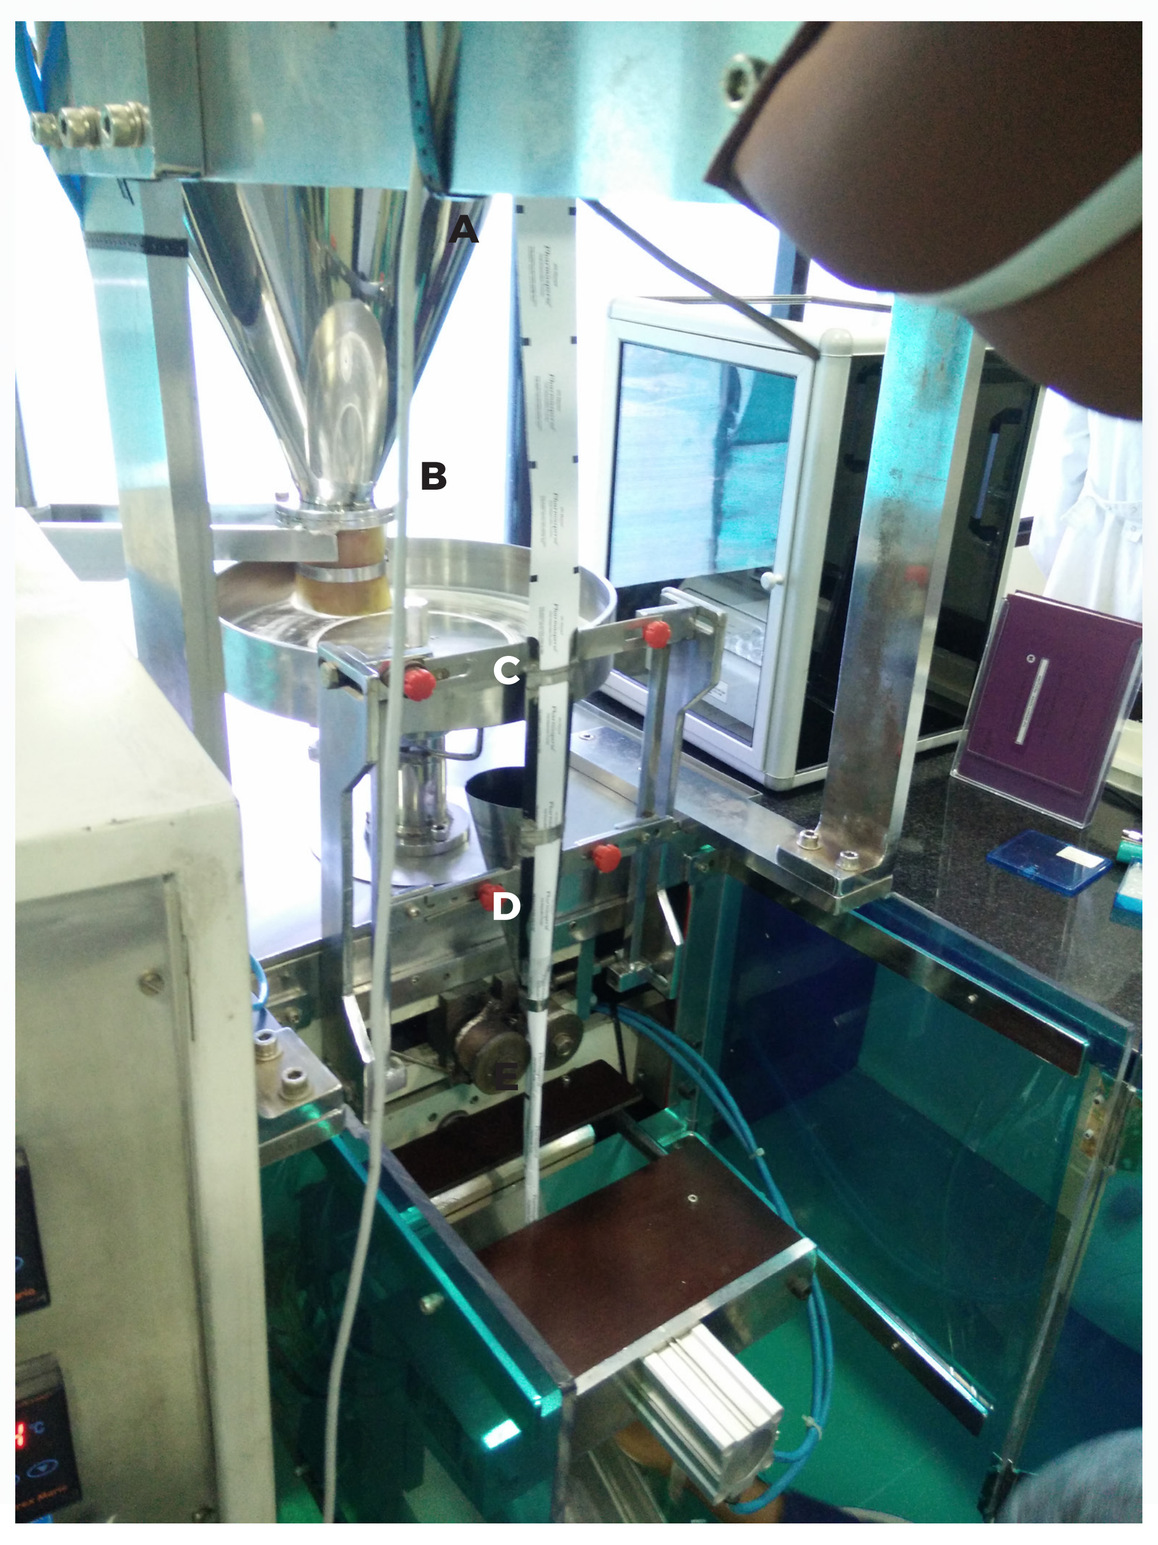

A
B
C
D

Hopper (A) contains the powder blend that relies on gravity feeding into four dies at (B). (C) The blend in each die is dropped into a small cone shaped hopper and flows into the empty stickpack sachet. At (D) Each stickpack is sealed vertically at 180˚C. At (E) each stick pack is further sealed horizontally at 80˚C and cut into the final stickpack.

Figure 2. Configuration of Hopper and Die That Control the Stickpack Fill Weight by Gravity Feeding

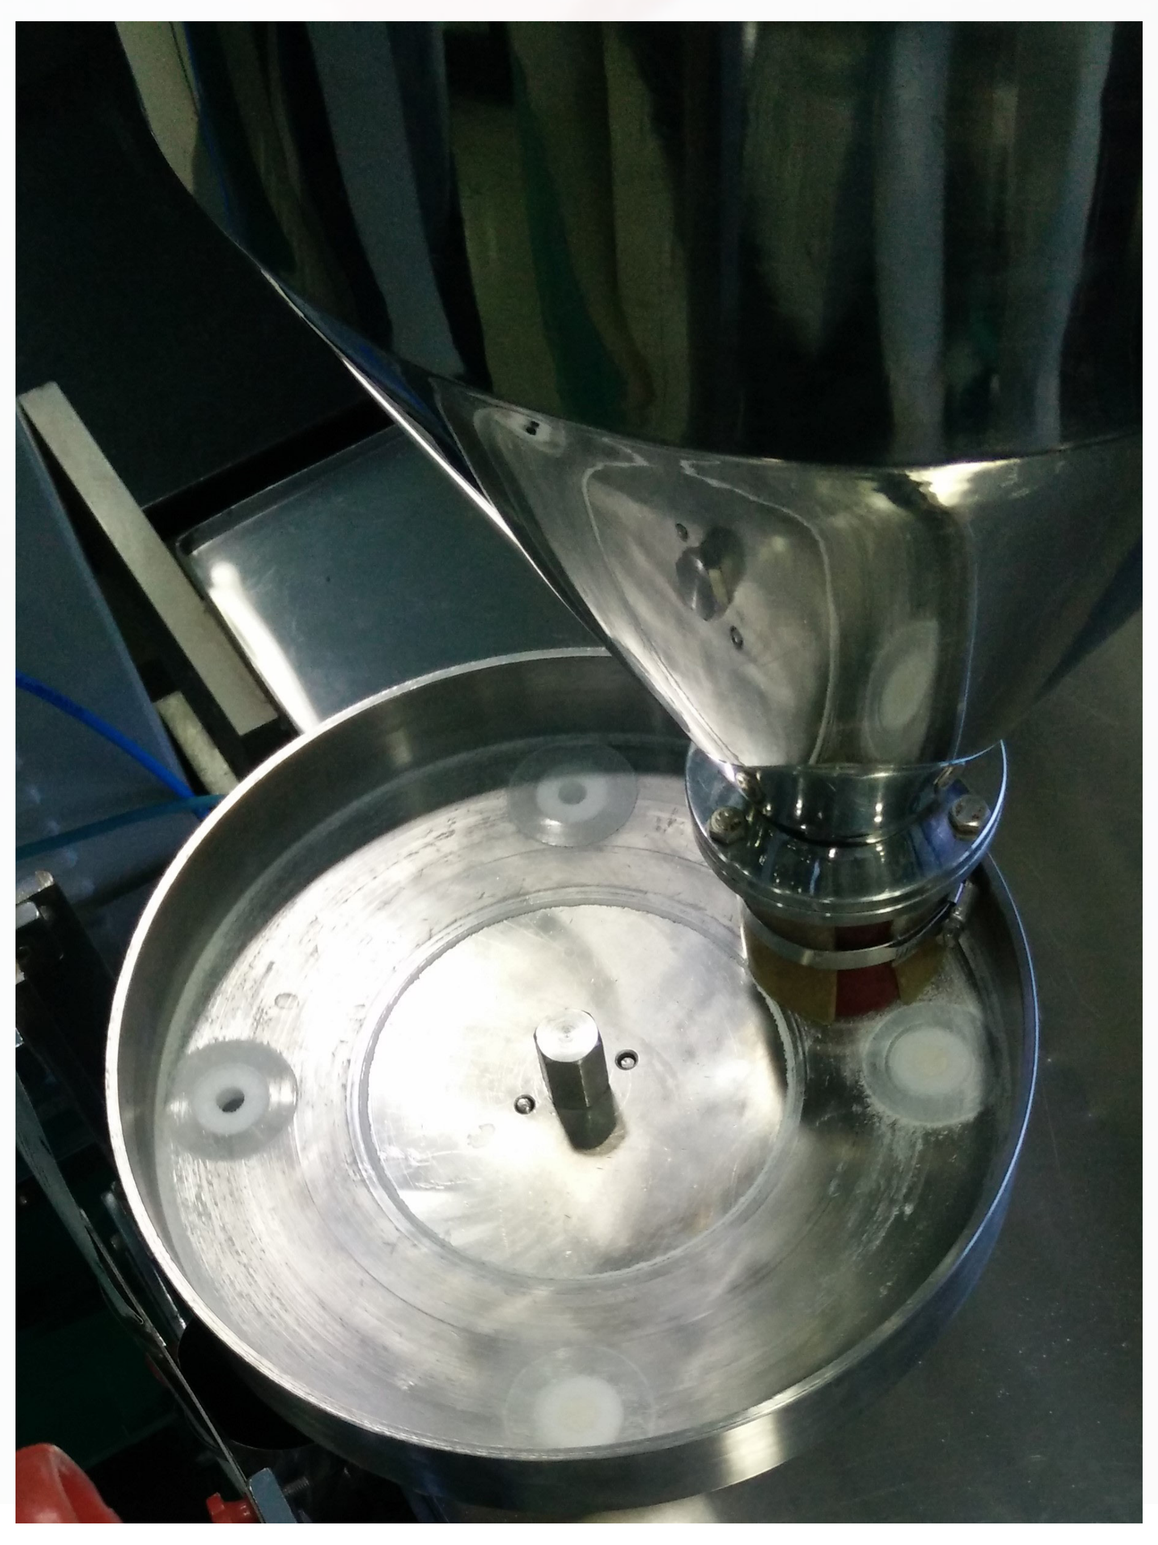

Figure 3. Dissolution of Paracetamol From the 4 ODP Formulations Studied

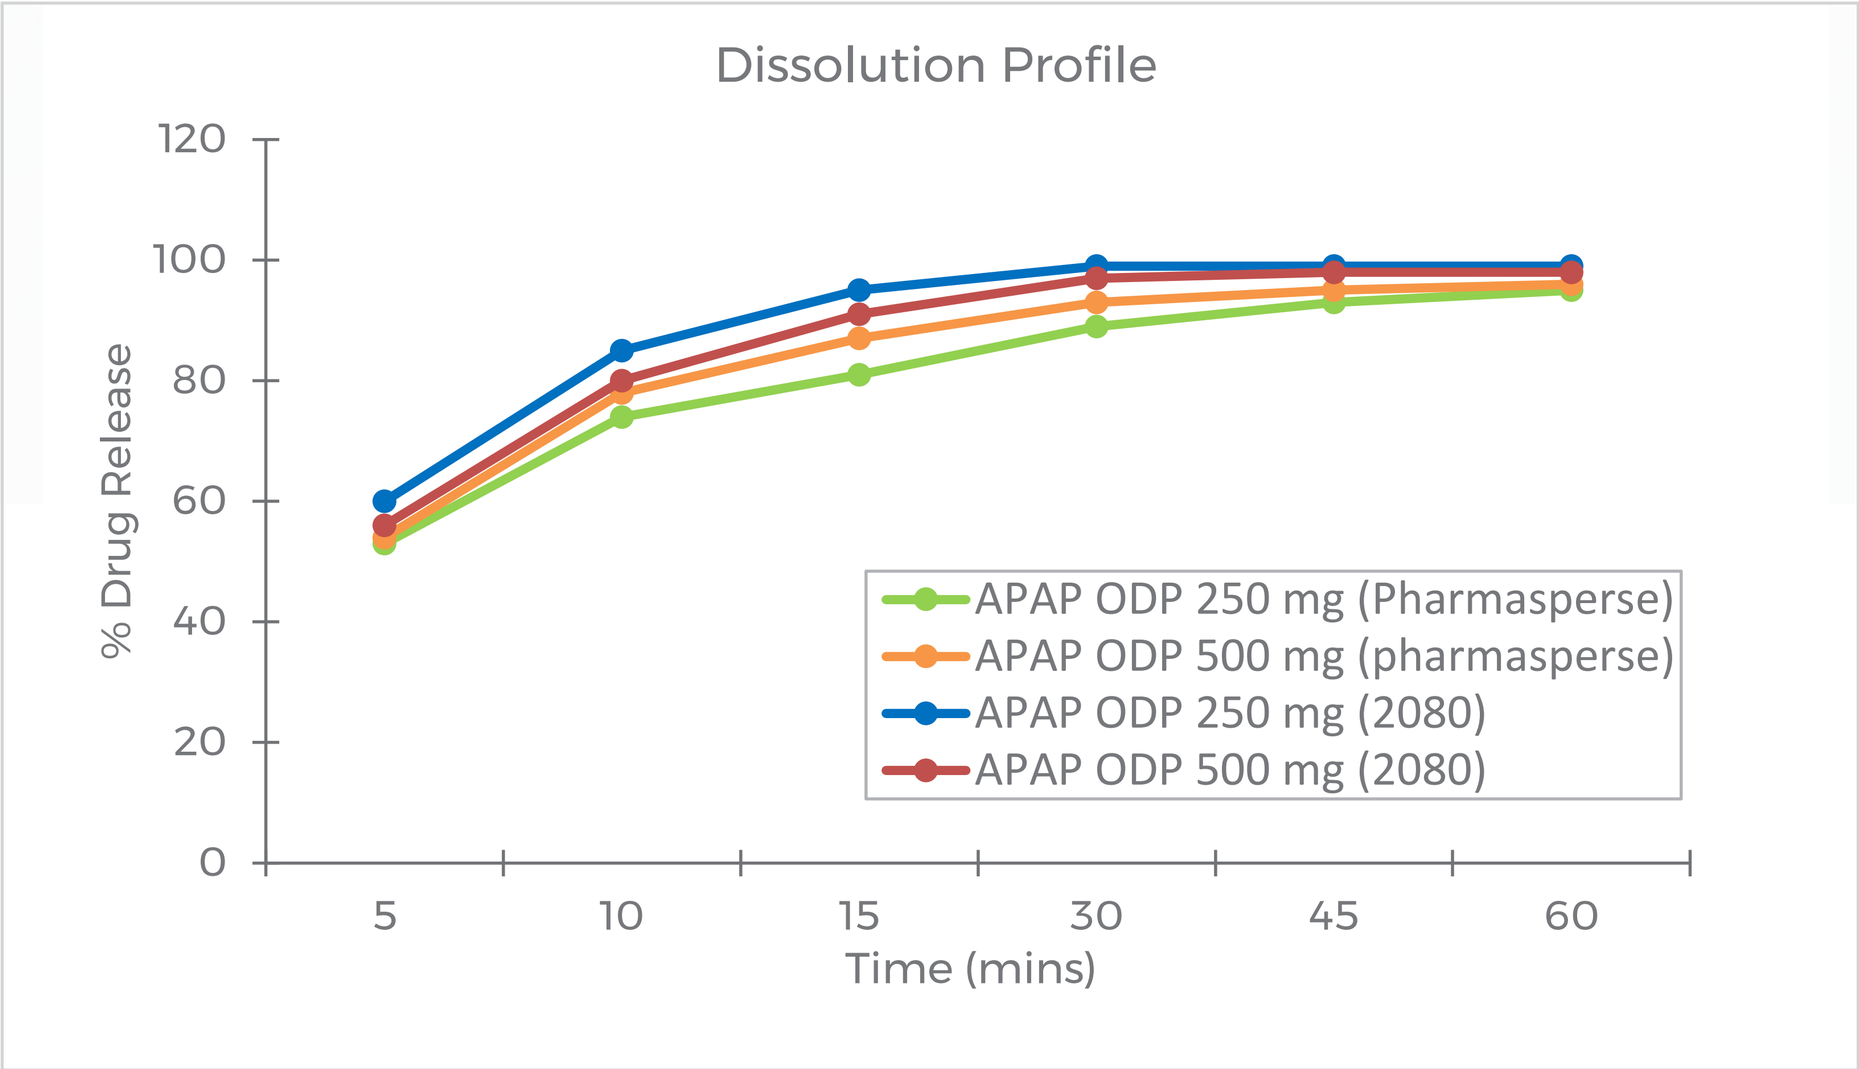

Dissolution Profile
% Drug Release
120
100
80
60
40
20
0
5
10
15
30
45
60
Time (mins)
APAP ODP 250 mg (Pharmasperse)
APAP ODP 500 mg (pharmasperse)
APAP ODP 250 mg (2080)
APAP ODP 500 mg (2080)

CONCLUSIONS

ODP formulation requires careful consideration of the excipient properties relative to the API taste to ensure a robust patient friendly dose form is developed. Key issues that influence the formulation robustness are the API particle size, PSD of the carrier material and drug loading level. Choice of carriers such as Pharmasperse that are engineered specifically for ODP formulation, greatly improve the patient experience via enhanced organoleptics and ease of use, benefiting the formulator and patient alike.

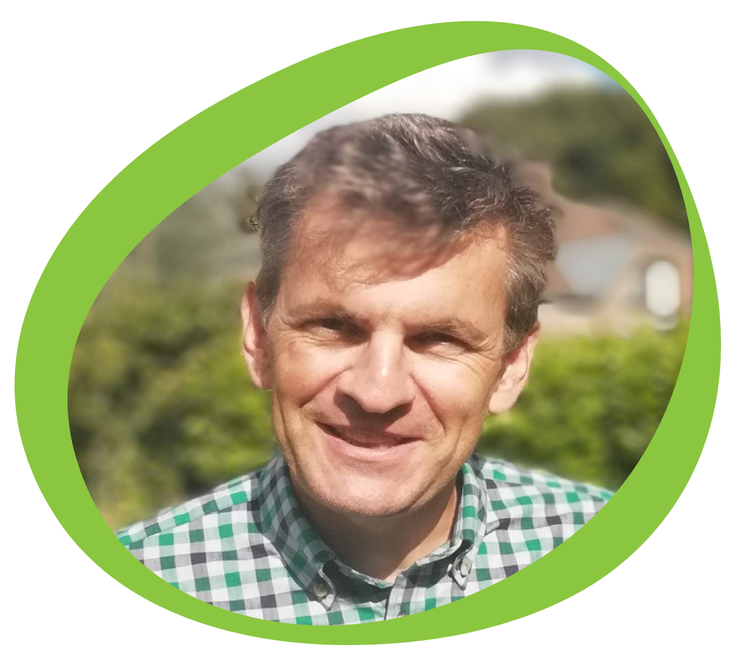

Graeme Macleod, PhD

Head of Global R&D

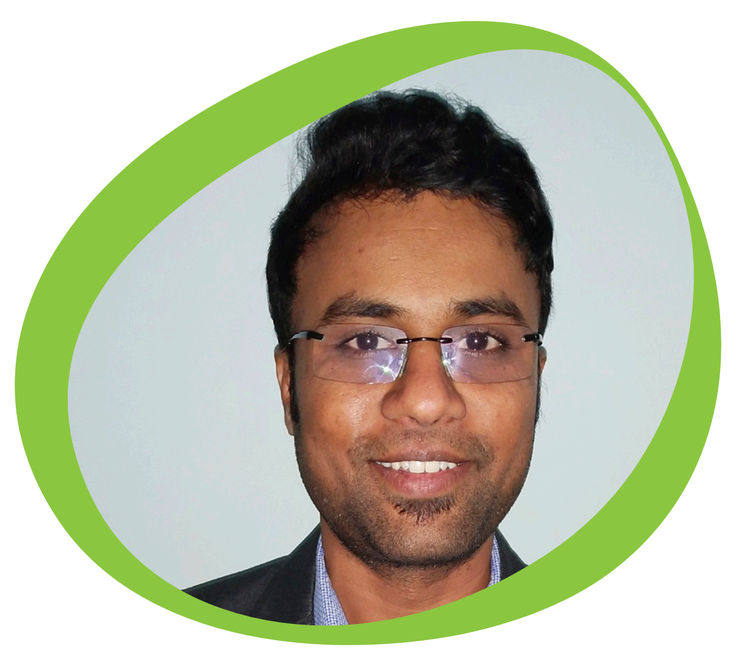

Hanumantharaja

Analyst

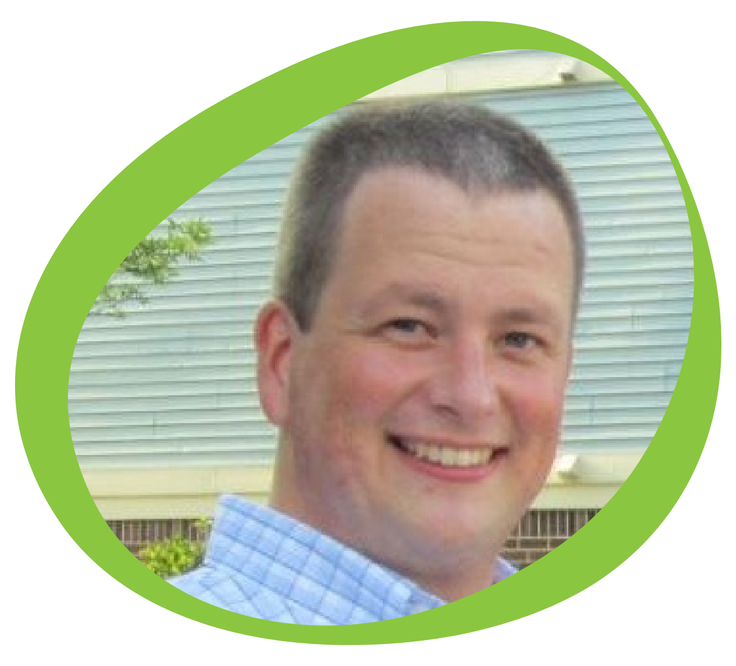

Brian Wilson, PhD

Global Applications, Research, & Technical Service Manager

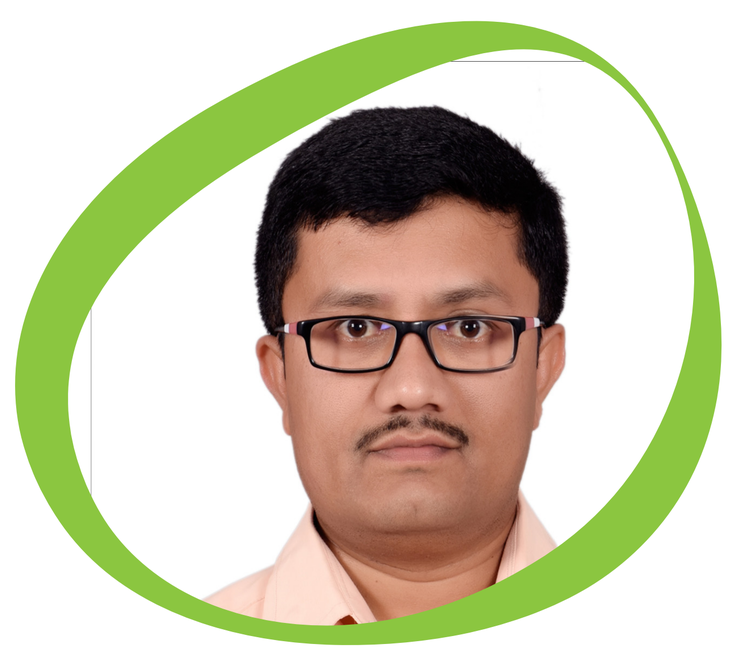

Girish BV, PhD

Group Leader Analytical Research

SPI Pharma™ Making a Difference

An ABF Ingredients Company